Extended preclinical ocular biodistribution and pharmacodynamic profile of ISTH0036, an antisense oligonucleotide targeting transforming growth factor beta 2 (TGF-β2) for the treatment of ophthalmic diseases

Presentation No.: 5702

WOSIKOWSKI, K.[1], KORBMACHER, B.[2], KRAMPERT, M.[3], ROEHL, I.[3], AND JANICOT, M.[1]

[1]Isarna Therapeutics GmbH, Munich, Germany, [2]Covance Preclinical Services, Münster, Germany, [3]Axolabs, Kulmbach, Germany

www.isarna-therapeutics.com
k.wosikowski@isarna-therapeutics.com

Abstract

Purpose: A critical role for TGF-β2 in the pathophysiology of several ocular diseases such as glaucoma, age-related macular degeneration or diabetic macular edema has been demonstrated, making this isoform a relevant therapeutic target. To support clinical development in these indications, the ocular biodistribution and pharmacodynamics profile of ISTH0036, a 14-mer phosphorothiate locked nucleic acid-modified antisense oligodeoxynucleotide gapmer, was evaluated in Cynomolgus monkeys following intravitreal administration.

Methods: To assess the time-dependent ocular tissue drug biodistribution and pharmacodynamics effects, ISTH0036 was administered (intravitreal injection) on Day 1 and Day 57 at a dose of 100 µg/eye into Cynomolgus monkey eyes. Furthermore, single administration of the compound at increasing doses of 30, 100 and 300 µg/eye was performed to assess the dose-dependency (on Day 29). An anion-exchange-HPLC method with fluorescence detection was used to analyze tissue concentrations. Target downregulation was determined using a branched DNA assay, and protein concentration in vitreous/aqueous humors was measured with a multiplex ELISA assay.

Results: Long-lasting and time-dependent biodistribution of ISTH0036 to the posterior eye tissues was observed. Similar high ISTH0036 concentrations were measured in the retina, choroid and ciliary body after 300-µg/eye administration (5-6 µg/g, on Day 29). High median drug concentrations in posterior eye tissues were still observed on the last day of measurement (Day 113). ISTH0036 induced *in vivo* long-lasting and dose-dependent TGF-β2 mRNA downregulation in retina and lens. TGF-β2 protein concentration decreased in the vitreous humor after intravitreal injection of ISTH0036 and this effect was maintained up to Day 113. These results confirm previous findings in the rabbit eye.

Conclusion: ISTH0036 demonstrated potent target TGF-β2 mRNA downregulation in relevant tissues of the Cynomolgus monkey eye. Pronounced long-lasting posterior eye tissue distribution was consistent with observed target engagement. Demonstrated biodistribution and target engagement support further clinical development and provide rationale for Q2M or Q3M administration schedule of ISTH0036 for therapeutic intervention in ophthalmology.

Study Design

Treatment: 1 or 2 intravitreal (IVT) injection(s), on Day 1 and Day 57 (group 3 only) in both eyes
Doses: 30, 100 and 300 µg/eye (corresponding to a calculated initial concentration in the vitreous humor C_0 ~ 3, 10 and 30 µM, respectively). Dosing volume was 25 µL
Tissue, vitreous and aqueous humor collection on Day 29 (group 1-4), Day 57 (Group 3), Day 85 (Group 3) and Day 113 (Group 3)
Group size: 1F, 1M monkey per group

Fig. 1: Study design

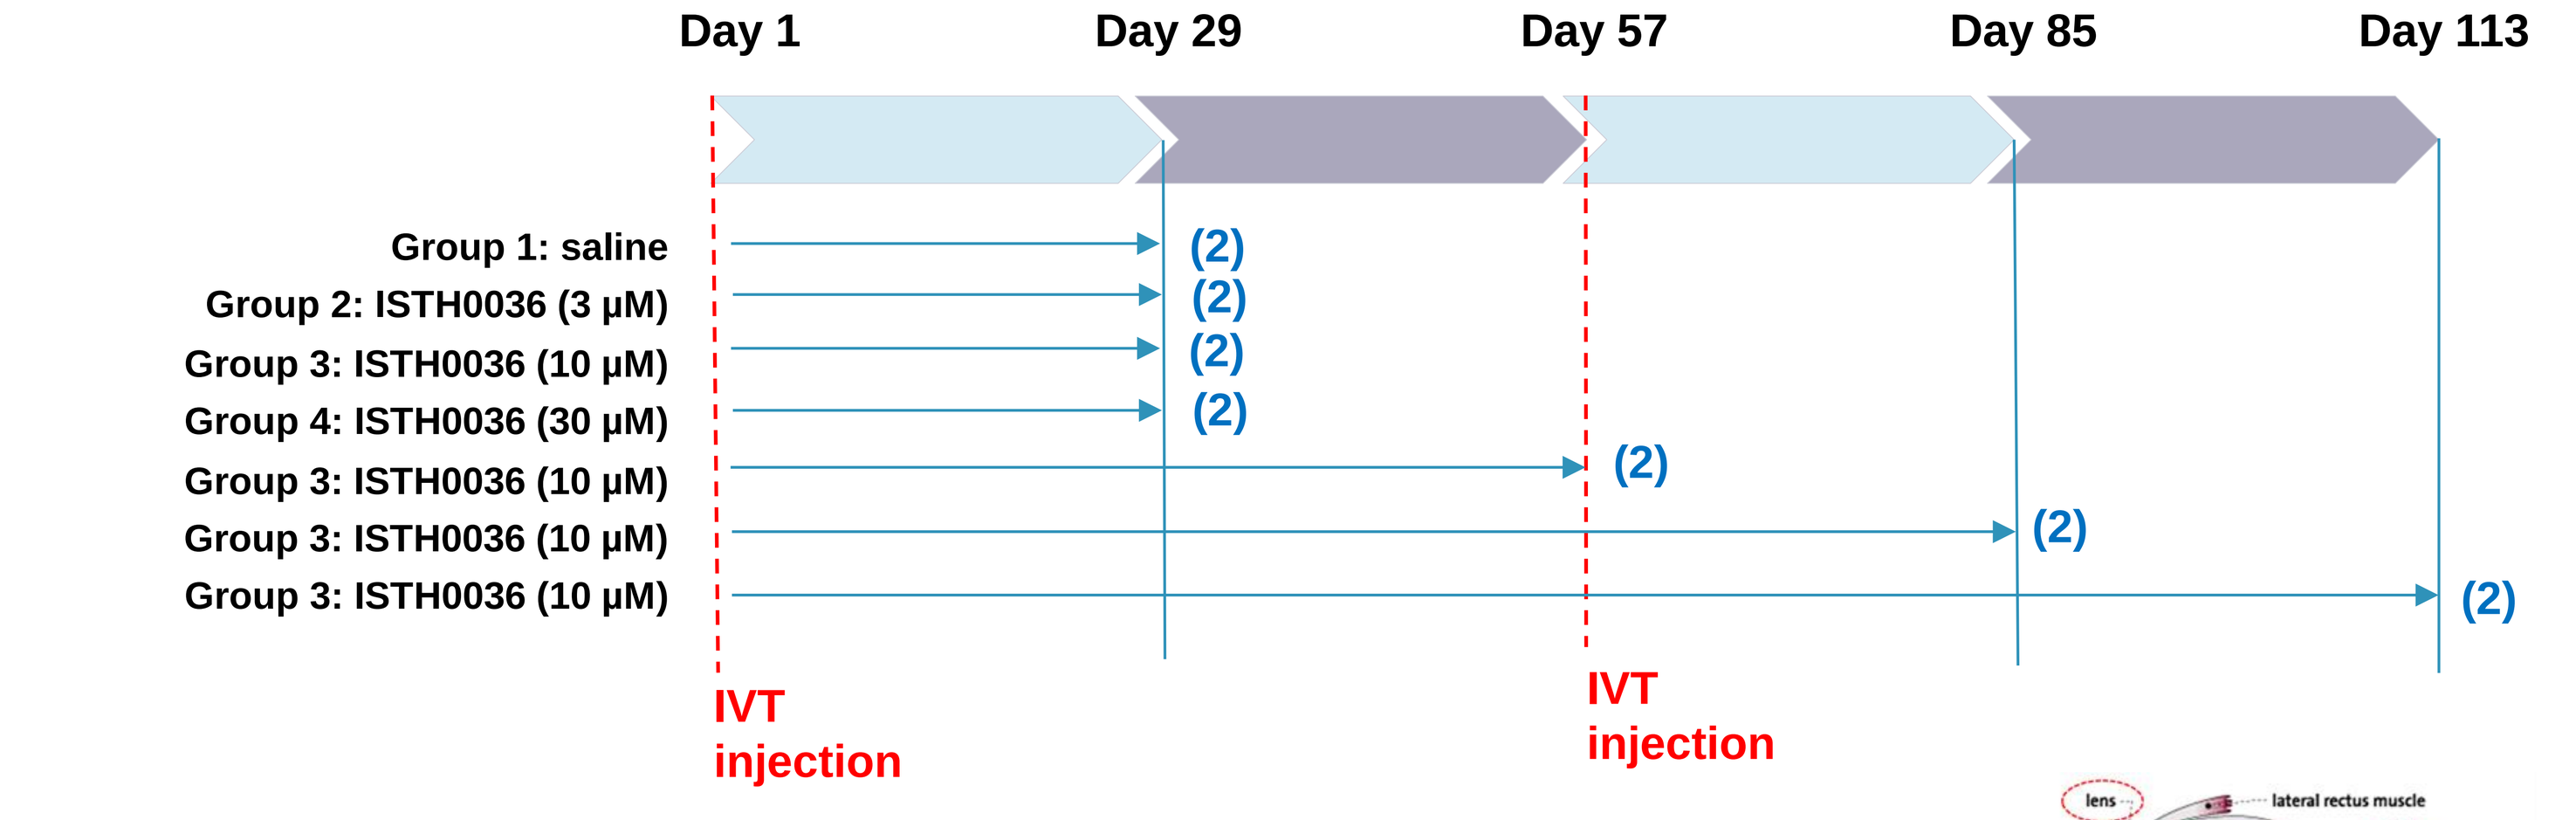

Day 1
Day 29
Day 57
Day 85
Day 113
Group 1: saline
Group 2: ISTH0036 (3 µM)
Group 3: ISTH0036 (10 µM)
Group 4: ISTH0036 (30 µM)
Group 3: ISTH0036 (10 µM)
Group 3: ISTH0036 (10 µM)
Group 3: ISTH0036 (10 µM)
(2)
IVT injection
IVT injection

	Drug concentration	Target engagement	TGF-beta multiplex
Aqueous and vitreous humor	-	-	☑
Ciliary Body	☑	☑	-
Lens	☑	☑	-
Retina	☑	☑	-
Choroid	☑	☑	-
Optic nerve head	☑	☑	-
Kidney cortex	☑	☑	-

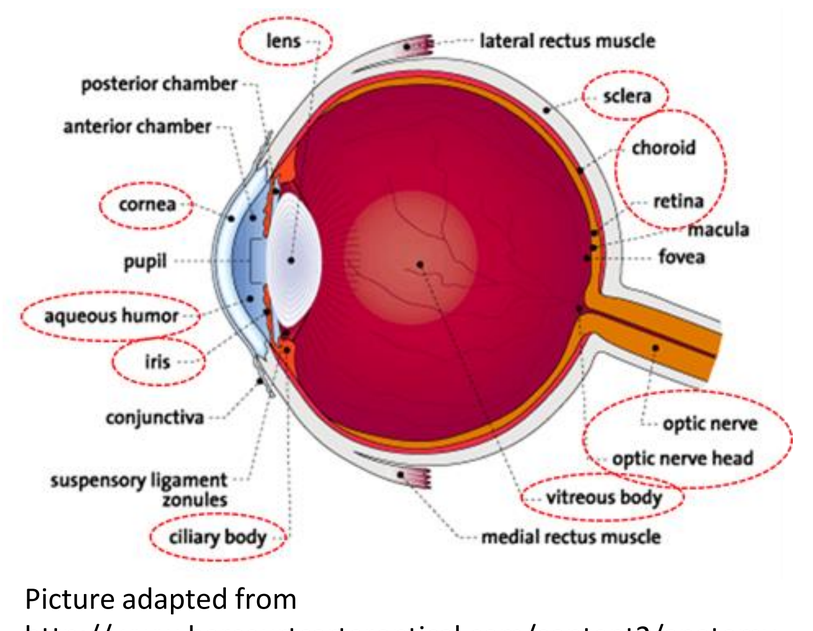

lens
lateral rectus muscle
posterior chamber
anterior chamber
sclera
choroid
cornea
retina
macula
fovea
pupil
aqueous humor
iris
conjunctiva
optic nerve
optic nerve head
suspensory ligament zonules
vitreous body
ciliary body
medial rectus muscle

Picture adapted from http://www.harcourtcarteroptical.com/content2/anatomy

The monkeys were treated at the Covance Preclinical Services GmbH test facility, which is fully accredited by the Association for Assessment and Accreditation of Laboratory Animal Care. All procedures in the study complied with the German Animal Welfare Act and were approved by the local Institutional Animal Care and Use Committee.

Inhibition of TGF-β2 as Target for Multi-modal Effects in Ophthalmic Diseases

TGF-β2

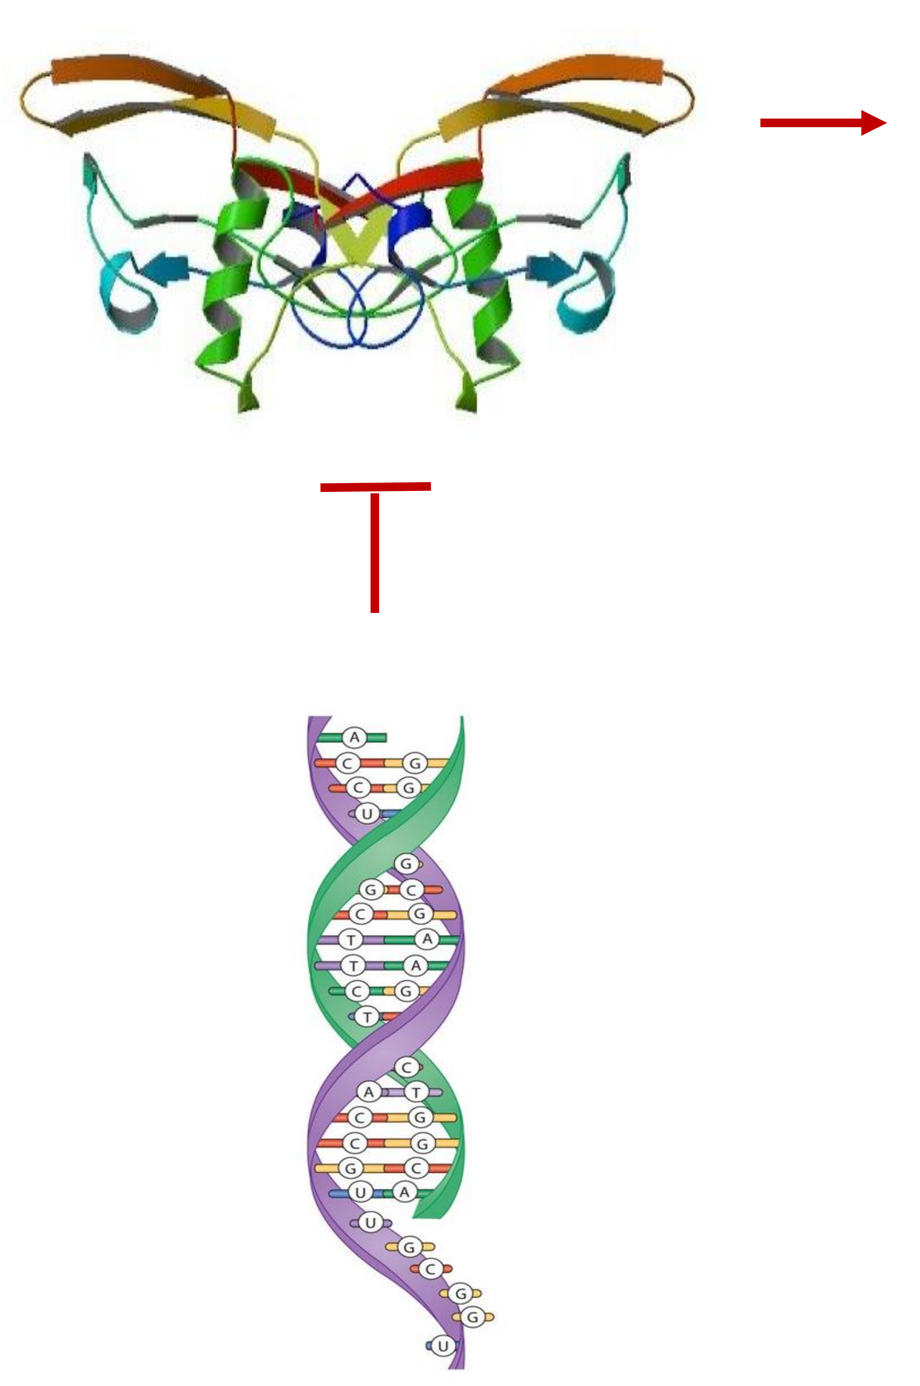

- One of the most important cytokines involved in the regulation of cell behavior in ocular tissues
- Predominant TGF-β isoform in the eye and found in large amounts in aqueous and vitreous humors and ocular tissues. Increased expression is reported in various ocular diseases (glaucoma, PVR, DR)
- Enhances gene expression related to tissue fibrosis, EMT, remodeling of ECM and inflammation
- Stimulates vascular endothelial cell proliferation and therefore a role in neovascularization is proposed
- Involved in optic nerve head remodeling and deformation of optic nerve axons

- Age-related macular degeneration
- Glaucoma
- Diabetic retinopathy
- Proliferative vitreoretinopathy

Compound

ISTH0036 is a fully phosphorothioate 14-mer oligodeoxynucleotide with a 3+3 LNA*-gapmer pattern selectively targeting TGF-β2 mRNA

5'- **GA**$^{(Me)}$**C**CAGATGCA**GGA** *-3'*

ISTH0036 Concentrations in Ocular Tissues and Kidney Cortex Cynomolgus Monkey Following IVT ISTH0036 Administration

Fig. 2A: Dose-dependency

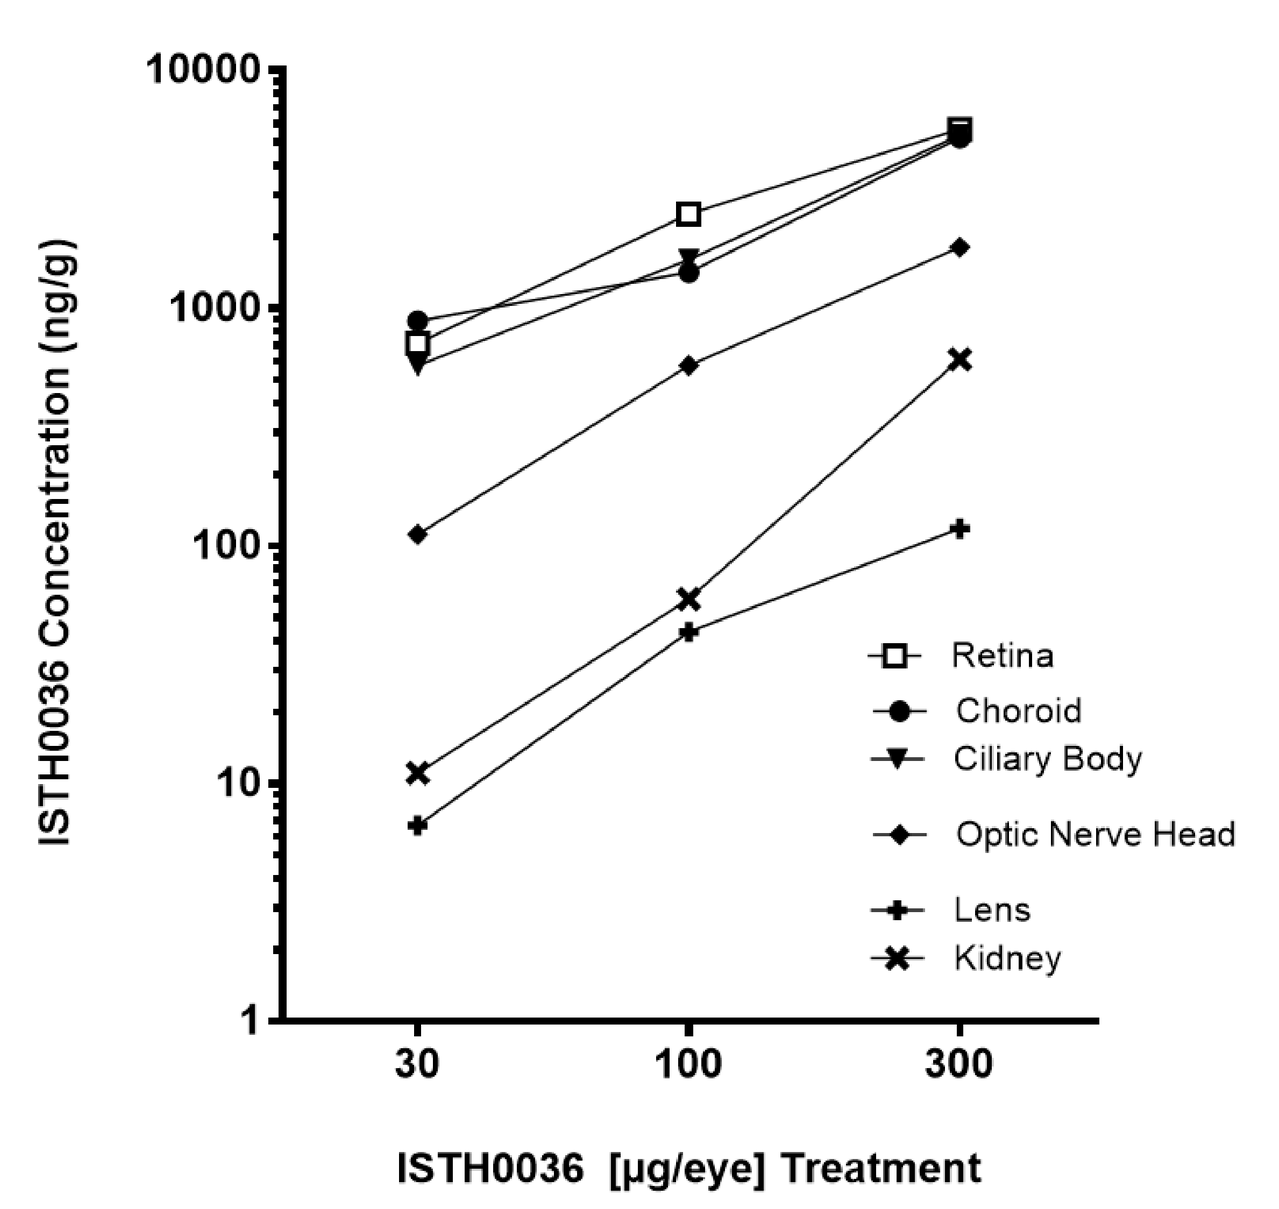

ISTH0036 Concentration (ng/g)
10000
1000
100
10
1
30
100
300
ISTH0036 [µg/eye] Treatment
Retina
Choroid
Ciliary Body
Optic Nerve Head
Lens
Kidney

Fig. 2B: Time-dependency

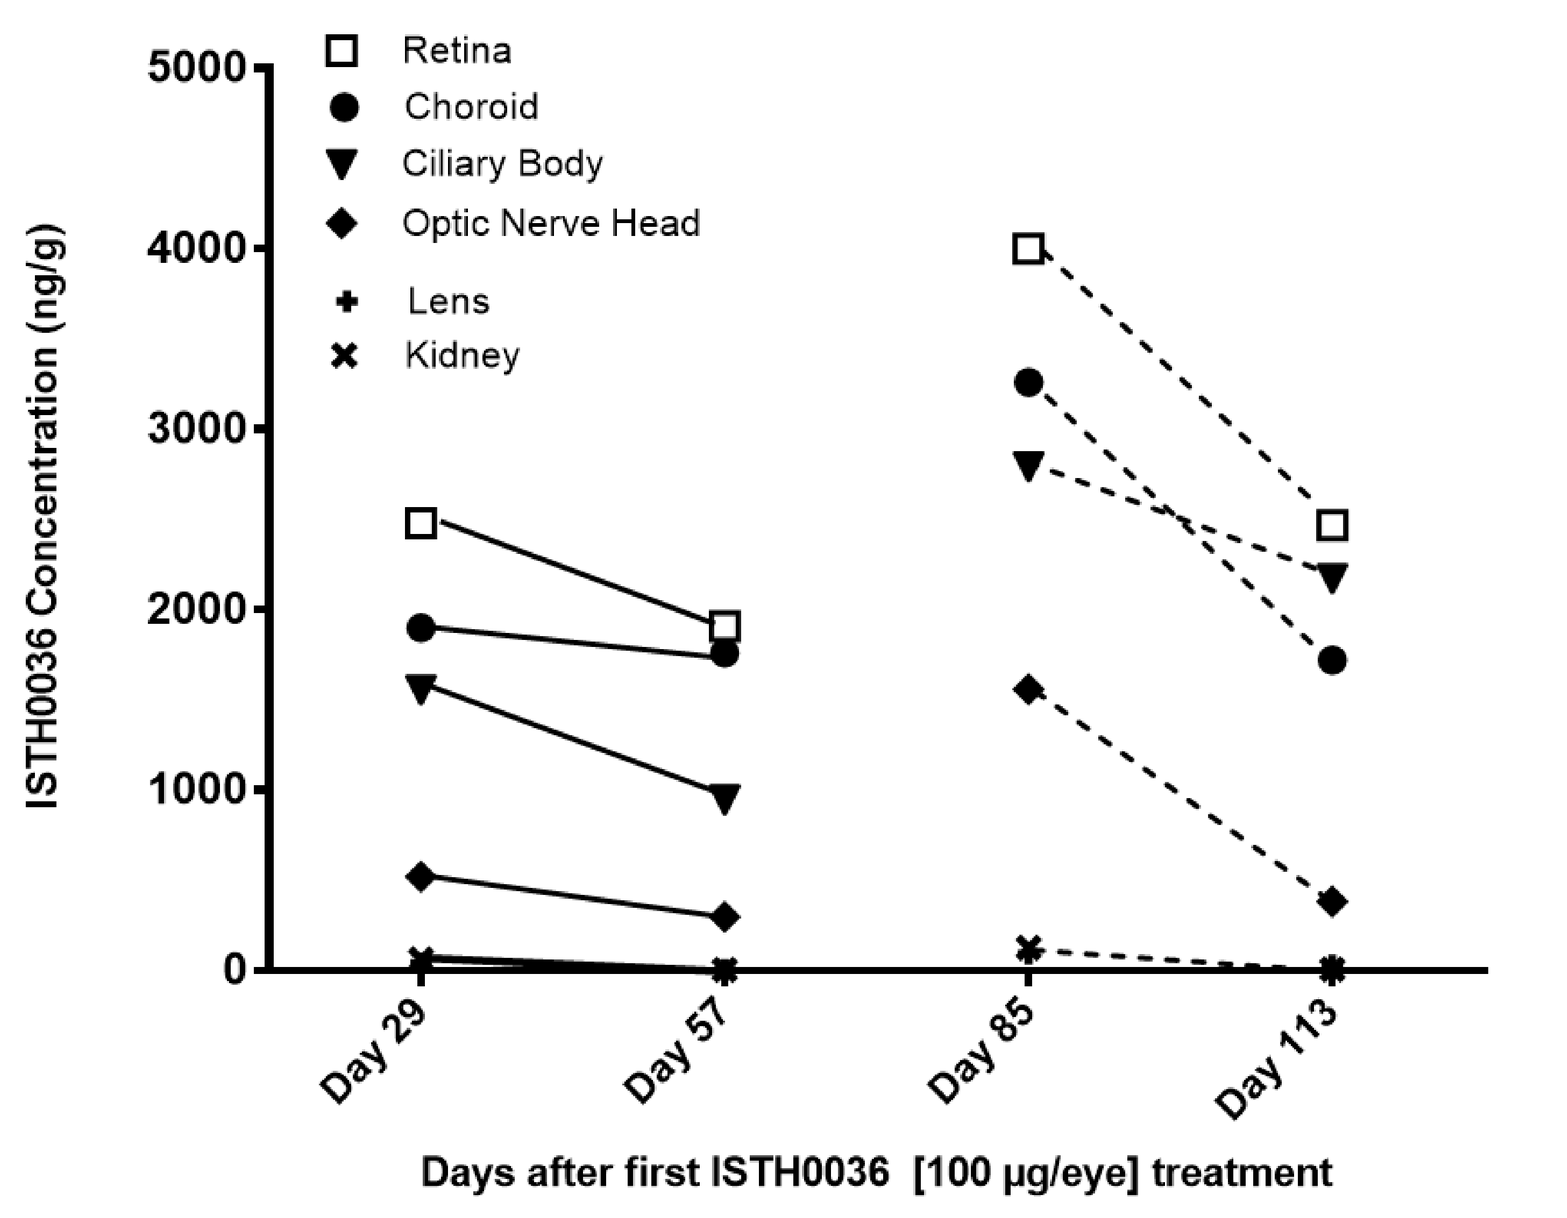

ISTH0036 Concentration (ng/g)
5000
4000
3000
2000
1000
0
Retina
Choroid
Ciliary Body
Optic Nerve Head
Lens
Kidney
Day 29
Day 57
Day 85
Day 113
Days after first ISTH0036 [100 µg/eye] treatment

Method: ISTH0036 was administered (IVT injection) to monkey eyes and ocular tissues collected according to the schema depicted in Fig. 1. An anion-exchange-HPLC method with fluorescence detection was used to analyze tissue concentrations. (**A**) ISTH0036 tissue concentrations after one IVT injection of ISTH0036 at 30, 100 or 300 µg/eye. Samples were collected on Day 29 after intraocular administration (n= 4 eyes, or kidney cortex samples from 2 animals). (**B**) ISTH0036 tissue concentrations after one injection on Day 1 (solid line) or two injections on Day 1 and 57 (dotted line) at 100 µg/eye. Samples were collected on Day 29, 57, 85 and 113 after the first intraocular administration (n= 4 eyes, or kidney cortex samples from 2 animals). Tissue biodistribution results are represented on (**A**) log- and (**B**) linear-scale as median ISTH0036 concentrations (n=4).

Results:

- Long-lasting and time-dependent drug distribution with a slow elimination rate in the examined posterior eye tissues
- Evidence of drug accumulation upon second administration
- Proportional dose-dependent drug distribution in all analyzed eye tissues
- Similar concentrations were reached in the retina, choroid, and ciliary body. The highest concentrations were measured in these tissues after 300 µg/eye administration (5-6 µg/g, on Day 29)
- Low drug concentrations in the kidney cortex (< 1 µg/g) following intraocular administration, suggesting some limited redistribution of ISTH0036 from the ocular compartment into the systemic blood circulation

Pharmacodynamic Profile in Ocular Tissues of Cynomolgus Monkey after IVT Injection

Fig. 3A: Dose-dependent target TGF-β2 mRNA downregulation in retina, lens and optic nerve head

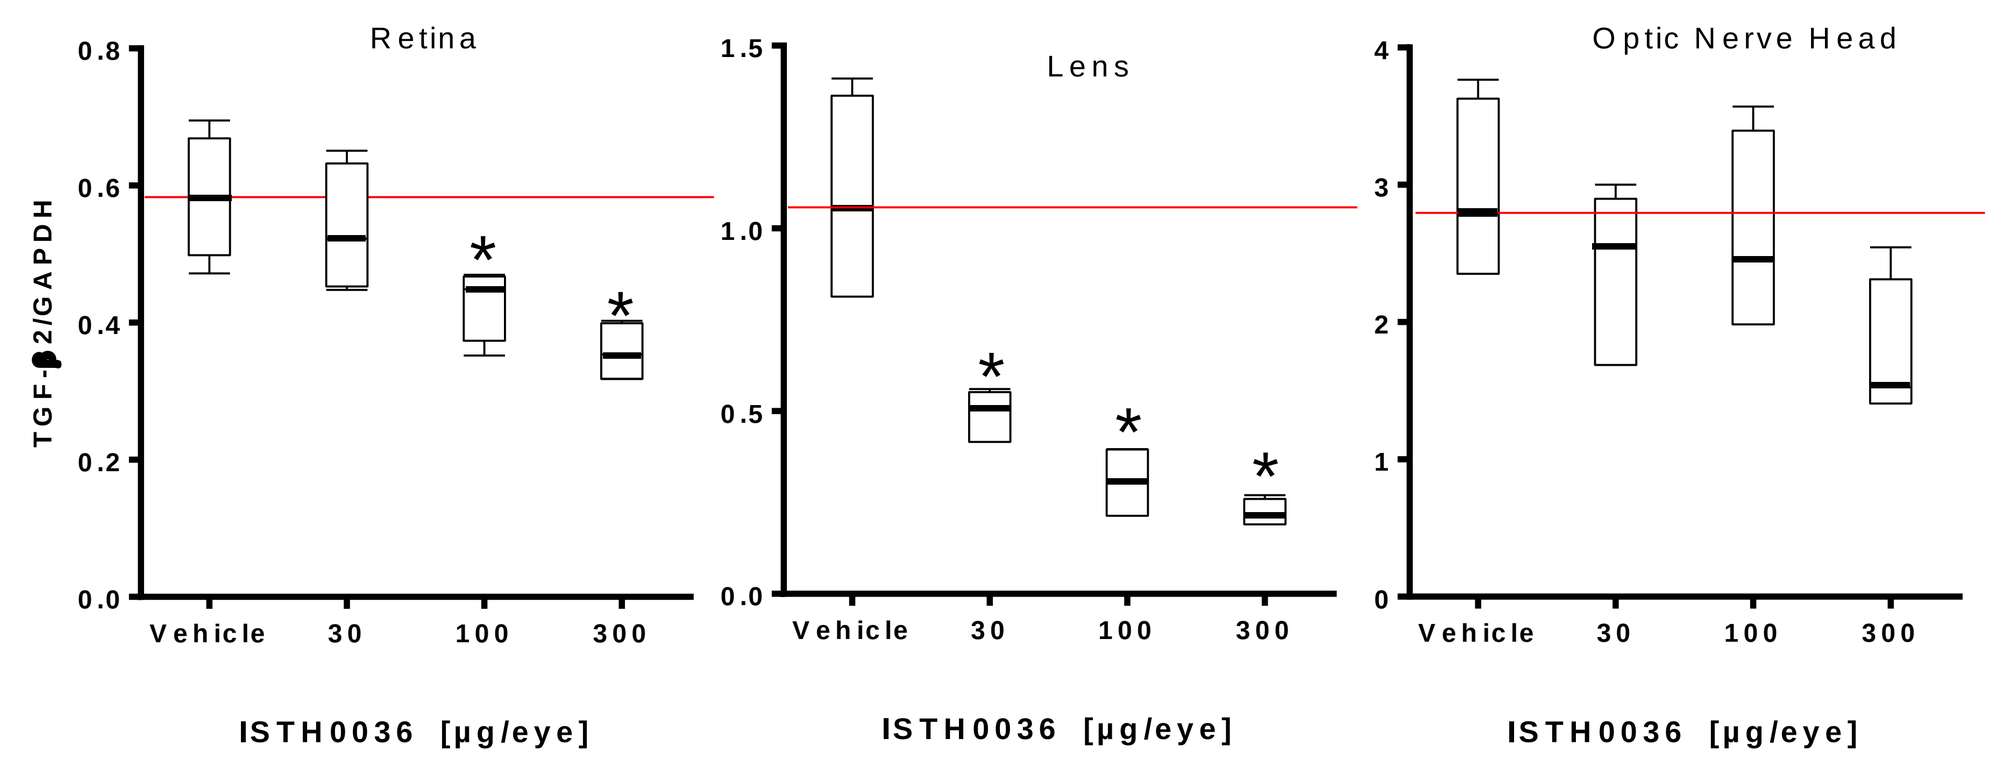

Retina
Lens
Optic Nerve Head
TGF-β2/GAPDH
Vehicle
30
100
300
ISTH0036 [µg/eye]

Fig. 3B: Time-dependent target TGF-β2 mRNA downregulation in retina, lens and optic nerve head

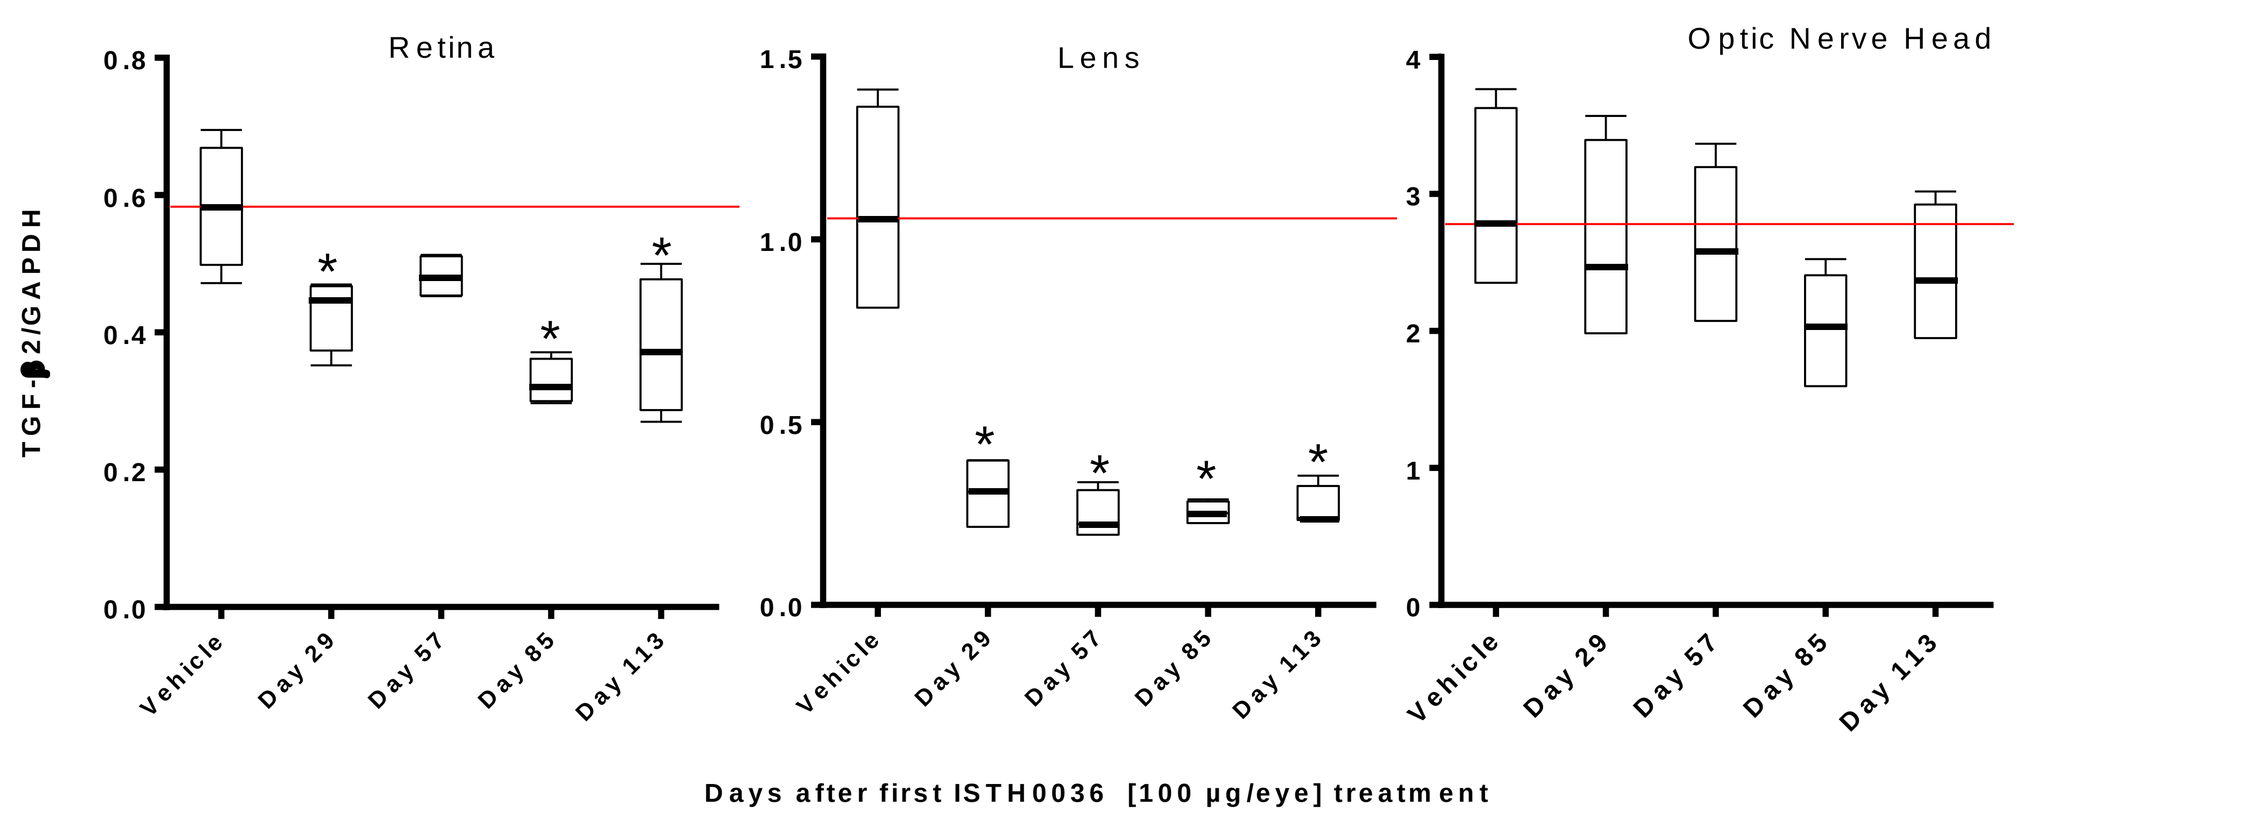

Retina
Lens
Optic Nerve Head
TGF-β2/GAPDH
Vehicle
Day 29
Day 57
Day 85
Day 113
Days after first ISTH0036 [100 µg/eye] treatment

Fig. 3C: Dose and time-dependent TGF-β2 protein downregulation in vitreous humor

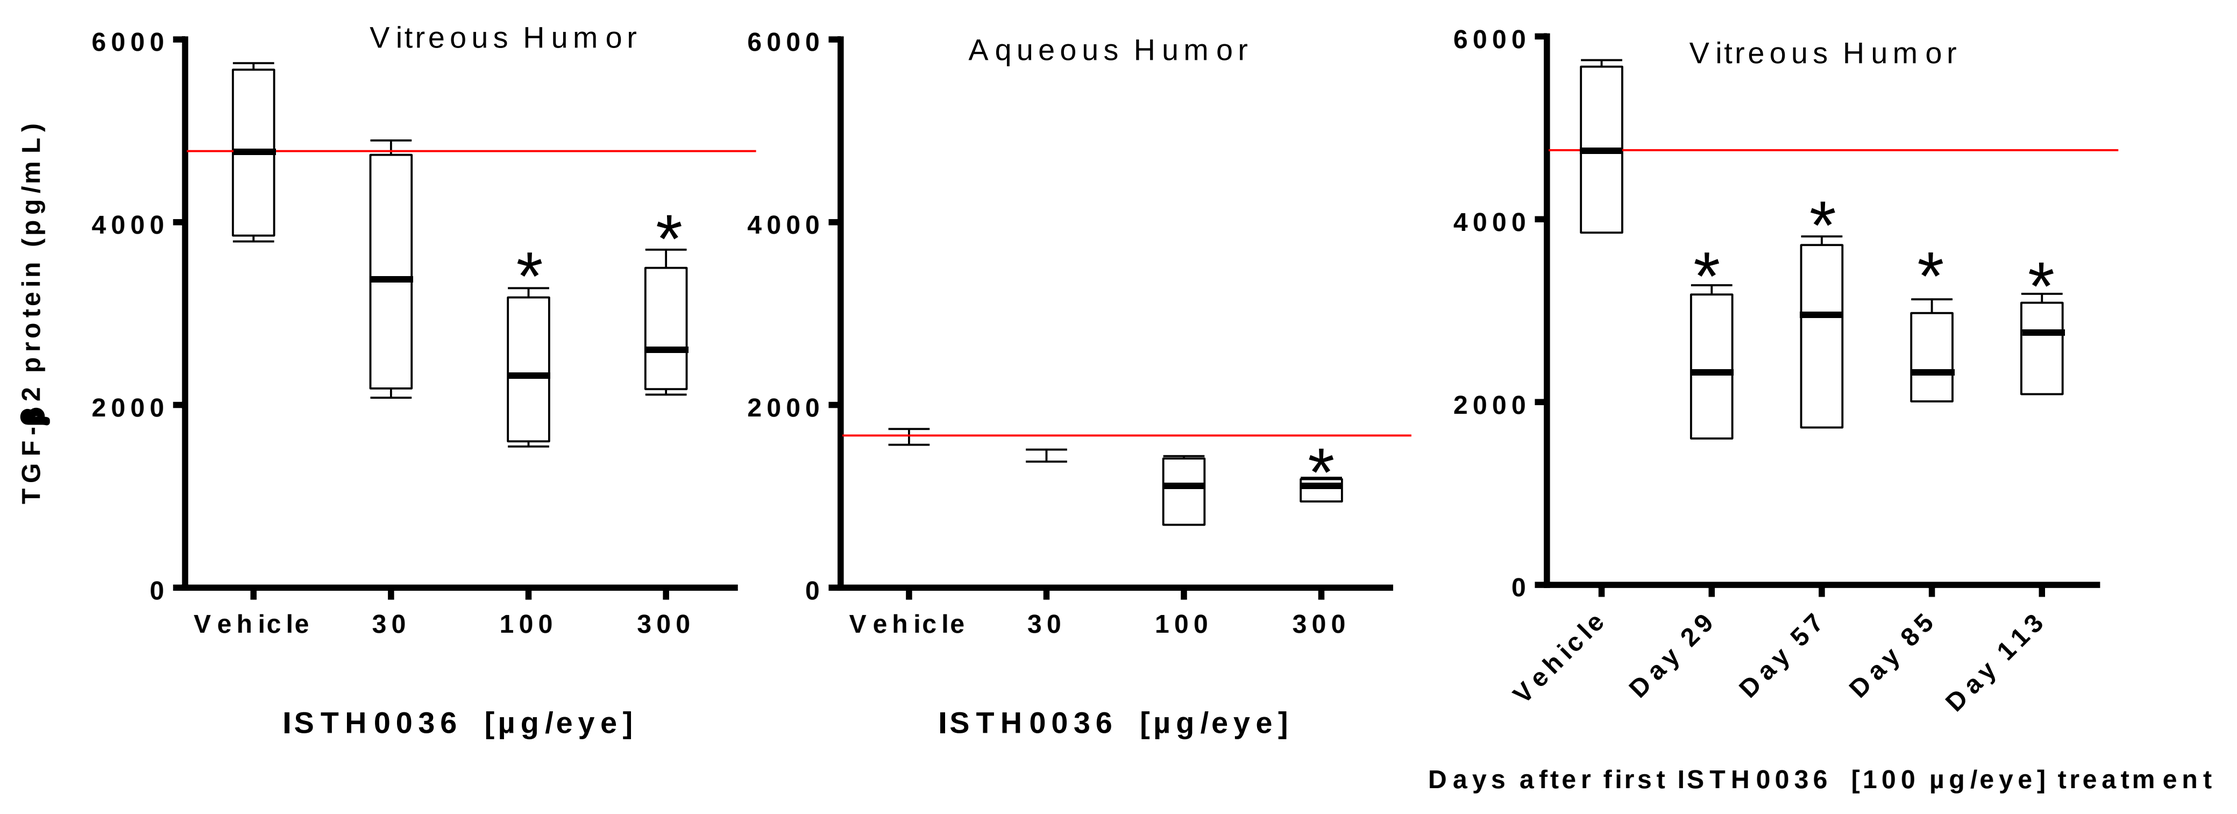

Vitreous Humor
Aqueous Humor
Vitreous Humor
TGF-β2 protein (pg/mL)
Vehicle
30
100
300
ISTH0036 [µg/eye]
Day 29
Day 57
Day 85
Day 113
Days after first ISTH0036 [100 µg/eye] treatment

Method: ISTH0036 was administered to monkey eyes via one or two IVT injection(s) and ocular tissues collected according to the schema depicted in Fig. 1. **(A+B)** Ocular tissues were dissected and immediately snap frozen for further analysis of target mRNA expression. TGF-β2 mRNA levels were quantified by bDNA assay and results were normalized to GAPDH values. **(C)** Aqueous and vitreous humor was collected and immediately snap frozen for further analysis of TGF-β1, -β2 and -β3 protein concentration. Protein levels were determined by electrochemiluminescence–based multiplex immuno-assay (U-PLEX TGF-β Combo NHP, Meso Scale Diagnostics LLC). TGF-β1 and -β3 protein levels were not detectable under given experimental procedures. Data are represented as box plots, in which median values (line), upper and lower quartiles, and 90th and 10th percentiles are indicated. * p<0.05 compared to vehicle treated group. Statistical significance was analyzed using non-parametric 2-independent samples Wilcoxon-Mann Whitney test.

Results:

- Significant time- and dose-dependent target engagement (TGF-β2 mRNA downregulation) in the retina and lens upon IVT administration(s) of ISTH0036
- Significant time- and dose-dependent target engagement (TGF-β2 protein expression) in vitreous humor. Minor effect in the aqueous humor

Conclusions

- Long lasting tissue distribution in ocular tissues after IVT administration, with minor systemic exposure as indicated by low exposure in kidney
- Long lasting, potent and selective *in vivo* target downregulation (TGF-β2 mRNA and protein)
- Data provide rationale for Q2M or Q3M administration schedule of ISTH0036 for therapeutic intervention in ophthalmology
- Data supportive of further clinical evaluation for treatment of patients with ocular diseases

*Use of LNA-modified gapmers is performed under a license from Roche.

ARVO Annual Meeting
Honolulu, April 29 - May 3, 2018